C041

Antitumor and Immunomodulatory Activity of ARV-6723, a PROTAC HPK1 Degrader, Across High- and Low-Immunogenic Solid Tumors

Anna C Van Acker*, William L Corwin*, Albert DeBerardinis, Emma Rousseau, Rebecca Conrad, Morena Scopel, Christopher Kuhlberg, Gregory Cadelina, Kim Davenport, Wendy Wu, John Corradi, Jennifer Pizzano, Keith R Hornberger, Angela M Cacace, Sean Landrette, XiaoZhe (Janet) Wang, Ignacio J Juncadella

Arvinas Operations, Inc., New Haven, CT, USA; *Co-first authors

Objectives

- To evaluate the preclinical antitumor activity of ARV-6723, a PROteolysis TArgeting Chimera (PROTAC) hematopoietic progenitor kinase 1 (HPK1) degrader, in syngeneic models of high- and low-immunogenic tumor microenvironments (TMEs), including an immune checkpoint inhibitor (ICI)-resistant pancreatic cancer model
- To compare the immunological changes induced by ARV-6723 vs a standard-of-care ICI and an investigational HPK1 inhibitor (HPK1i)

Key Findings

- ARV-6723 showed robust target engagement (HPK1 degradation), inhibition of a proximal pharmacodynamic biomarker (phosphorylation of SH2 domain-containing leukocyte protein of 76 kDa [SLP76]), and demonstrated single-agent antitumor activity in multiple syngeneic mouse tumor models, including high-immunogenic and low-immunogenic models
- Single-agent ARV-6723-driven tumor growth inhibition (TGI) surpassed that observed with an investigational HPK1i and an anti-programmed cell death protein 1 (PD-1) antibody in CT26, B16F10, and KPCY mouse models
- Immunological evaluation of tumor-infiltrating lymphocytes (TILs) revealed a distinct profile of proinflammatory changes (ie, increased infiltration of activated natural killer [NK] cells and monocytes, and decreased infiltration of immunosuppressive neutrophils) with ARV-6723 vs an investigational HPK1i
- RNA sequencing (RNA-seq) and pathway biomarker analyses provided mechanistic insight into the antitumor activity, suggesting that ARV-6723 elicits changes in proinflammatory pathways that are not observed with HPK1 inhibition

Conclusions

- ARV-6723 monotherapy demonstrated robust single-agent antitumor and proinflammatory activity in multiple syngeneic tumor models, including those with immunosuppressive TMEs, and showed greater preclinical activity than an investigational HPK1i or an anti-PD-1 antibody
- Immunoprofiling results suggest that the superior antitumor efficacy of ARV-6723 compared with HPK1 inhibition is driven by distinct immunomodulatory effects, likely resulting from sustained HPK1 degradation that targets both its kinase-dependent and kinase-independent functions
- These findings support clinical investigation of ARV-6723 monotherapy in patients with solid tumors harboring high- or low-immunogenic TMEs, including ICI-resistant tumor settings

References

1. Hu MC, et al. Genes Dev. 1996;10:2251-64.
2. Sawasdikosol S, Burakoff S. eLife 2020;9:e55122.
3. Lious J, et al. Immunity. 2000;12(4):399–408.
4. Ling P, et al. J Biol Chem. 2001;276(22):18908–14.
5. Alzabin S, et al. J Immunol. 2009;182(10):6187–94.
6. Liu J, et al. PLoS ONE. 2019;14(3):e0212670.
7. You D, et al. J Immunother Cancer. 2021;9(1):e001402.
8. Bader A, et al. J Immunol. 2022;52(5):760-769.
9. Choi WS, et al. Adv Sci. 2024;11(29):e2400920.
10. Seto T, et al. Méd Sci. 2019;7(2):14.
11. Memon D, et al. Cancer Cell. 2024;42(2):209-224.e9.
12. Song Y, et al. Bioorg Med Chem. 2026;134:118546.
13. Corwin W, et al. Poster presented at SITC 2025; #1158.
14. Yu JW, et al. PLoS One. 2018;13(11):e0206223.
15. Li J, et al. Immunity. 2018;49.1:178-193.

Acknowledgments

This research was funded by Arvinas, Inc. Medical writing support was provided by Lela Creutz, of Arvinas Operations, Inc. We thank Ben Z Stanger for KPCY cell lines[15] and insights into maintaining them.

AACR Immuno-Oncology (AACR IO)
Los Angeles, CA, USA; February 18–21, 2026

Background

- HPK1 (encoded by *MAP4K1*) is a serine/threonine protein kinase that is expressed almost exclusively in the hematopoietic compartment[1,2]
- This kinase has a well-defined role as a negative regulator of T-cell receptor signaling that acts proximally to the receptor and has been linked to decreased T-cell activation and proliferation and increased T-cell dysfunction and exhaustion[3,4]
- HPK1 also has negative regulatory roles in other immune cells (eg, B cells, NK cells, and dendritic cells [DCs]) through less well understood kinase-independent mechanisms[5-9]
- Although immuno-oncology therapies are effective in multiple solid-tumor types, substantial unmet medical need persists, as resistance mechanisms (eg, immunosuppressive TMEs) often contribute to disease progression[10,11]
- ARV-6723 is a potent and selective orally bioavailable PROTAC HPK1 degrader that will be investigated as a potential immunotherapy for the treatment of adult patients with advanced solid tumors[12]
- The PROTAC mechanism of action (**Figure 1**) and associated iterative event-driven activity may offer advantages over a small-molecule HPK1i because:
 - HPK1 kinase-independent functions and proposed scaffolding functions[12] are removed by protein degradation but not by kinase inhibition
 - Degradation of HPK1 allows for more durable pathway engagement than an occupancy-driven inhibitor mechanism
- In vitro, ARV-6723 demonstrated potent reduction of HPK1 levels and robust pathway engagement (inhibition of phosphorylation of SLP76), T-cell activation (interleukin-2), NK-cell activation (interferon-γ), and reversal of T-cell exhaustion[13]
- We previously showed robust antitumor activity of ARV-6723 in multiple syngeneic mouse models, with ARV-6723 demonstrating greater TGI than single-agent treatment with an anti-PD-1 antibody[13]
- Combining ARV-6723 with anti-PD-1 therapy elicited complete tumor regressions and immune memory in vivo[13]

Figure 1: Mechanism of action of ARV-6723

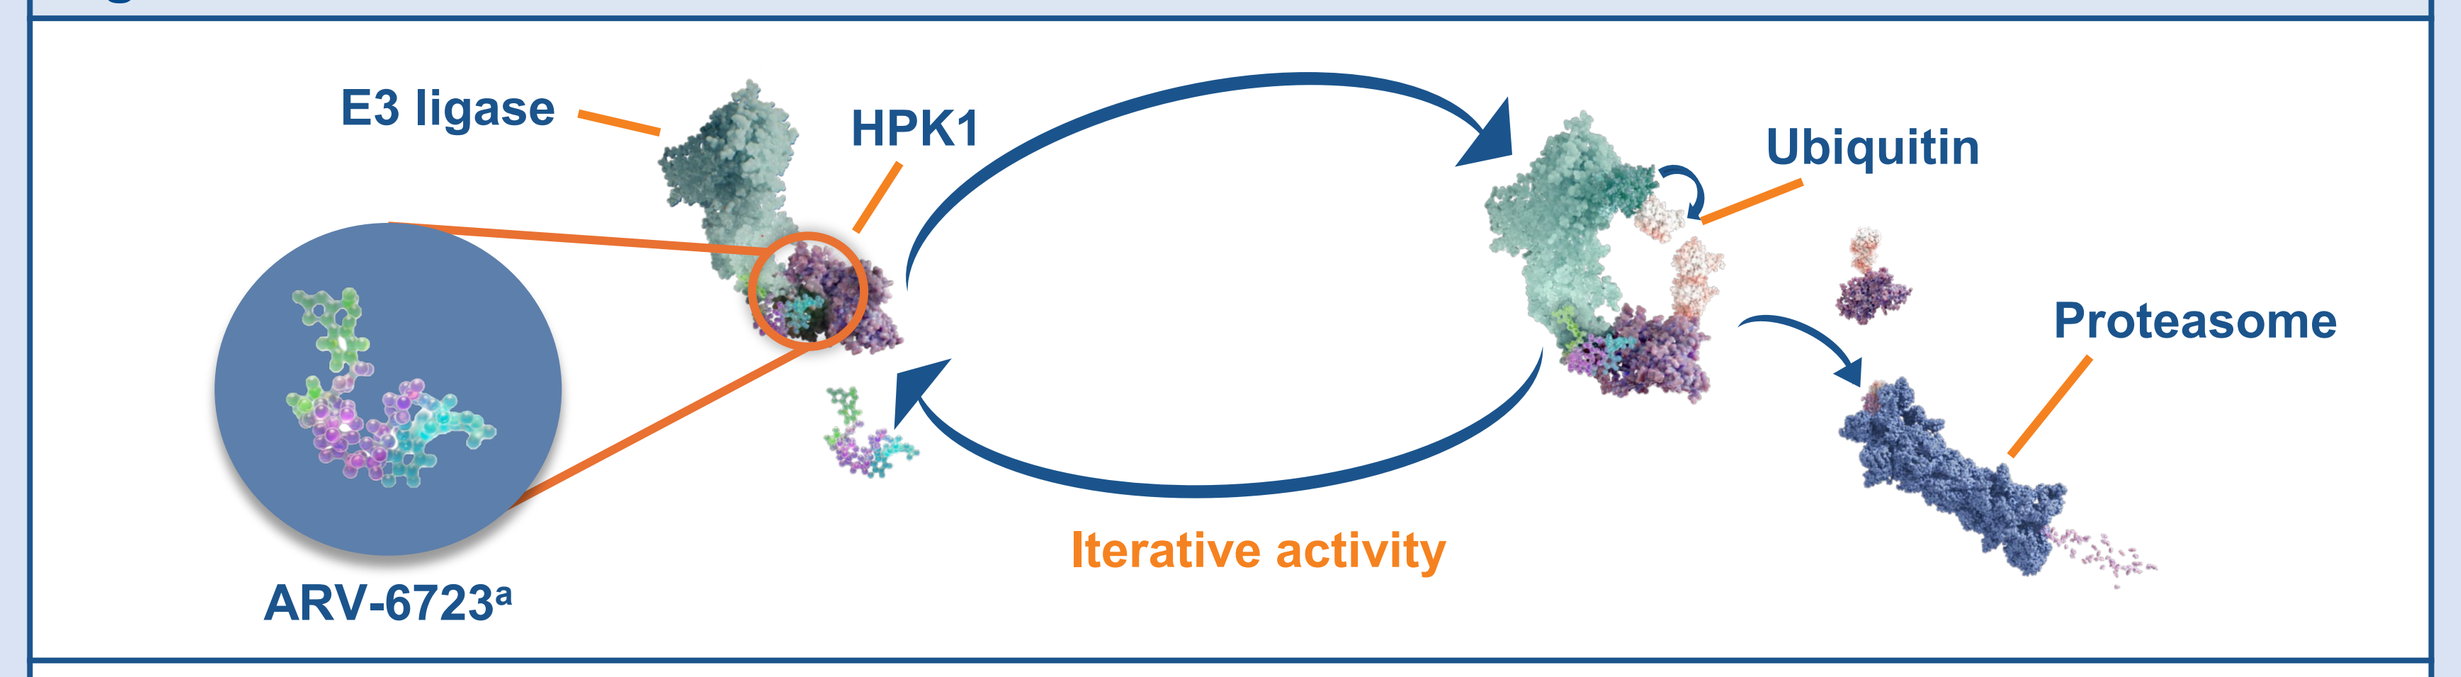

E3 ligase
HPK1
Ubiquitin
Proteasome
ARV-6723ᵃ
Iterative activity

ᵃGeneral PROTAC protein degrader is shown.
HPK1=hematopoietic progenitor kinase 1.

Results

- Single-agent ARV-6723 demonstrated deep HPK1 degradation and significant TGI that was superior to that observed with an anti-PD-1 antibody across a range of immunogenic syngeneic tumor models (**Figure 2**)
- ARV-6723 showed head-to-head superiority vs an investigational HPK1i in a low-immunogenic model (B16F10; TGI: 80% vs 2%; **Figure 3**) and a high-immunogenic model (CT26; TGI: 76% vs 15%; **Figure 4**)
- Flow cytometric immunophenotyping of TILs in the B16F10 model revealed distinct changes following ARV-6723 treatment, including reductions in neutrophil levels and increases in NK cells and Ly6C[hi] monocytes (**Figure 3**)
- Immune-cell deconvolution analysis of CT26 tumor-derived bulk RNA-seq data showed that ARV-6723 increased inferred proportions of proinflammatory cell types (activated NK cells, $CD8^+$ T cells, M1 macrophages, and memory B cells) and decreased endothelial fraction, neutrophils, and naive B cells (**Figure 4**)
- Bulk RNA-seq and gene enrichment analyses of CT26 and B16F10 tumors revealed gene expression changes unique to ARV-6723 treatment that were significantly associated with proinflammatory pathways and positive immunotherapy response signatures, suggesting that proinflammatory transcriptional changes underpin the enhanced ARV-6723 antitumor response in both models (**Figure 5**)
- Single-agent ARV-6723 demonstrated increased antitumor efficacy in an anti-PD-1-resistant pancreatic cancer model (KPCY) vs an investigational HPK1i (TGI: 89% vs 27%; **Figure 6A**)
- Single-nuclear RNA-seq analyses suggested that ARV-6723 uniquely induced proinflammatory pathways in macrophages (**Figure 6B**)

Figure 2: TGI and HPK1 degradation in a range of immunogenic syngeneic tumor models treated with ARV-6723 vs an anti-PD-1 antibody

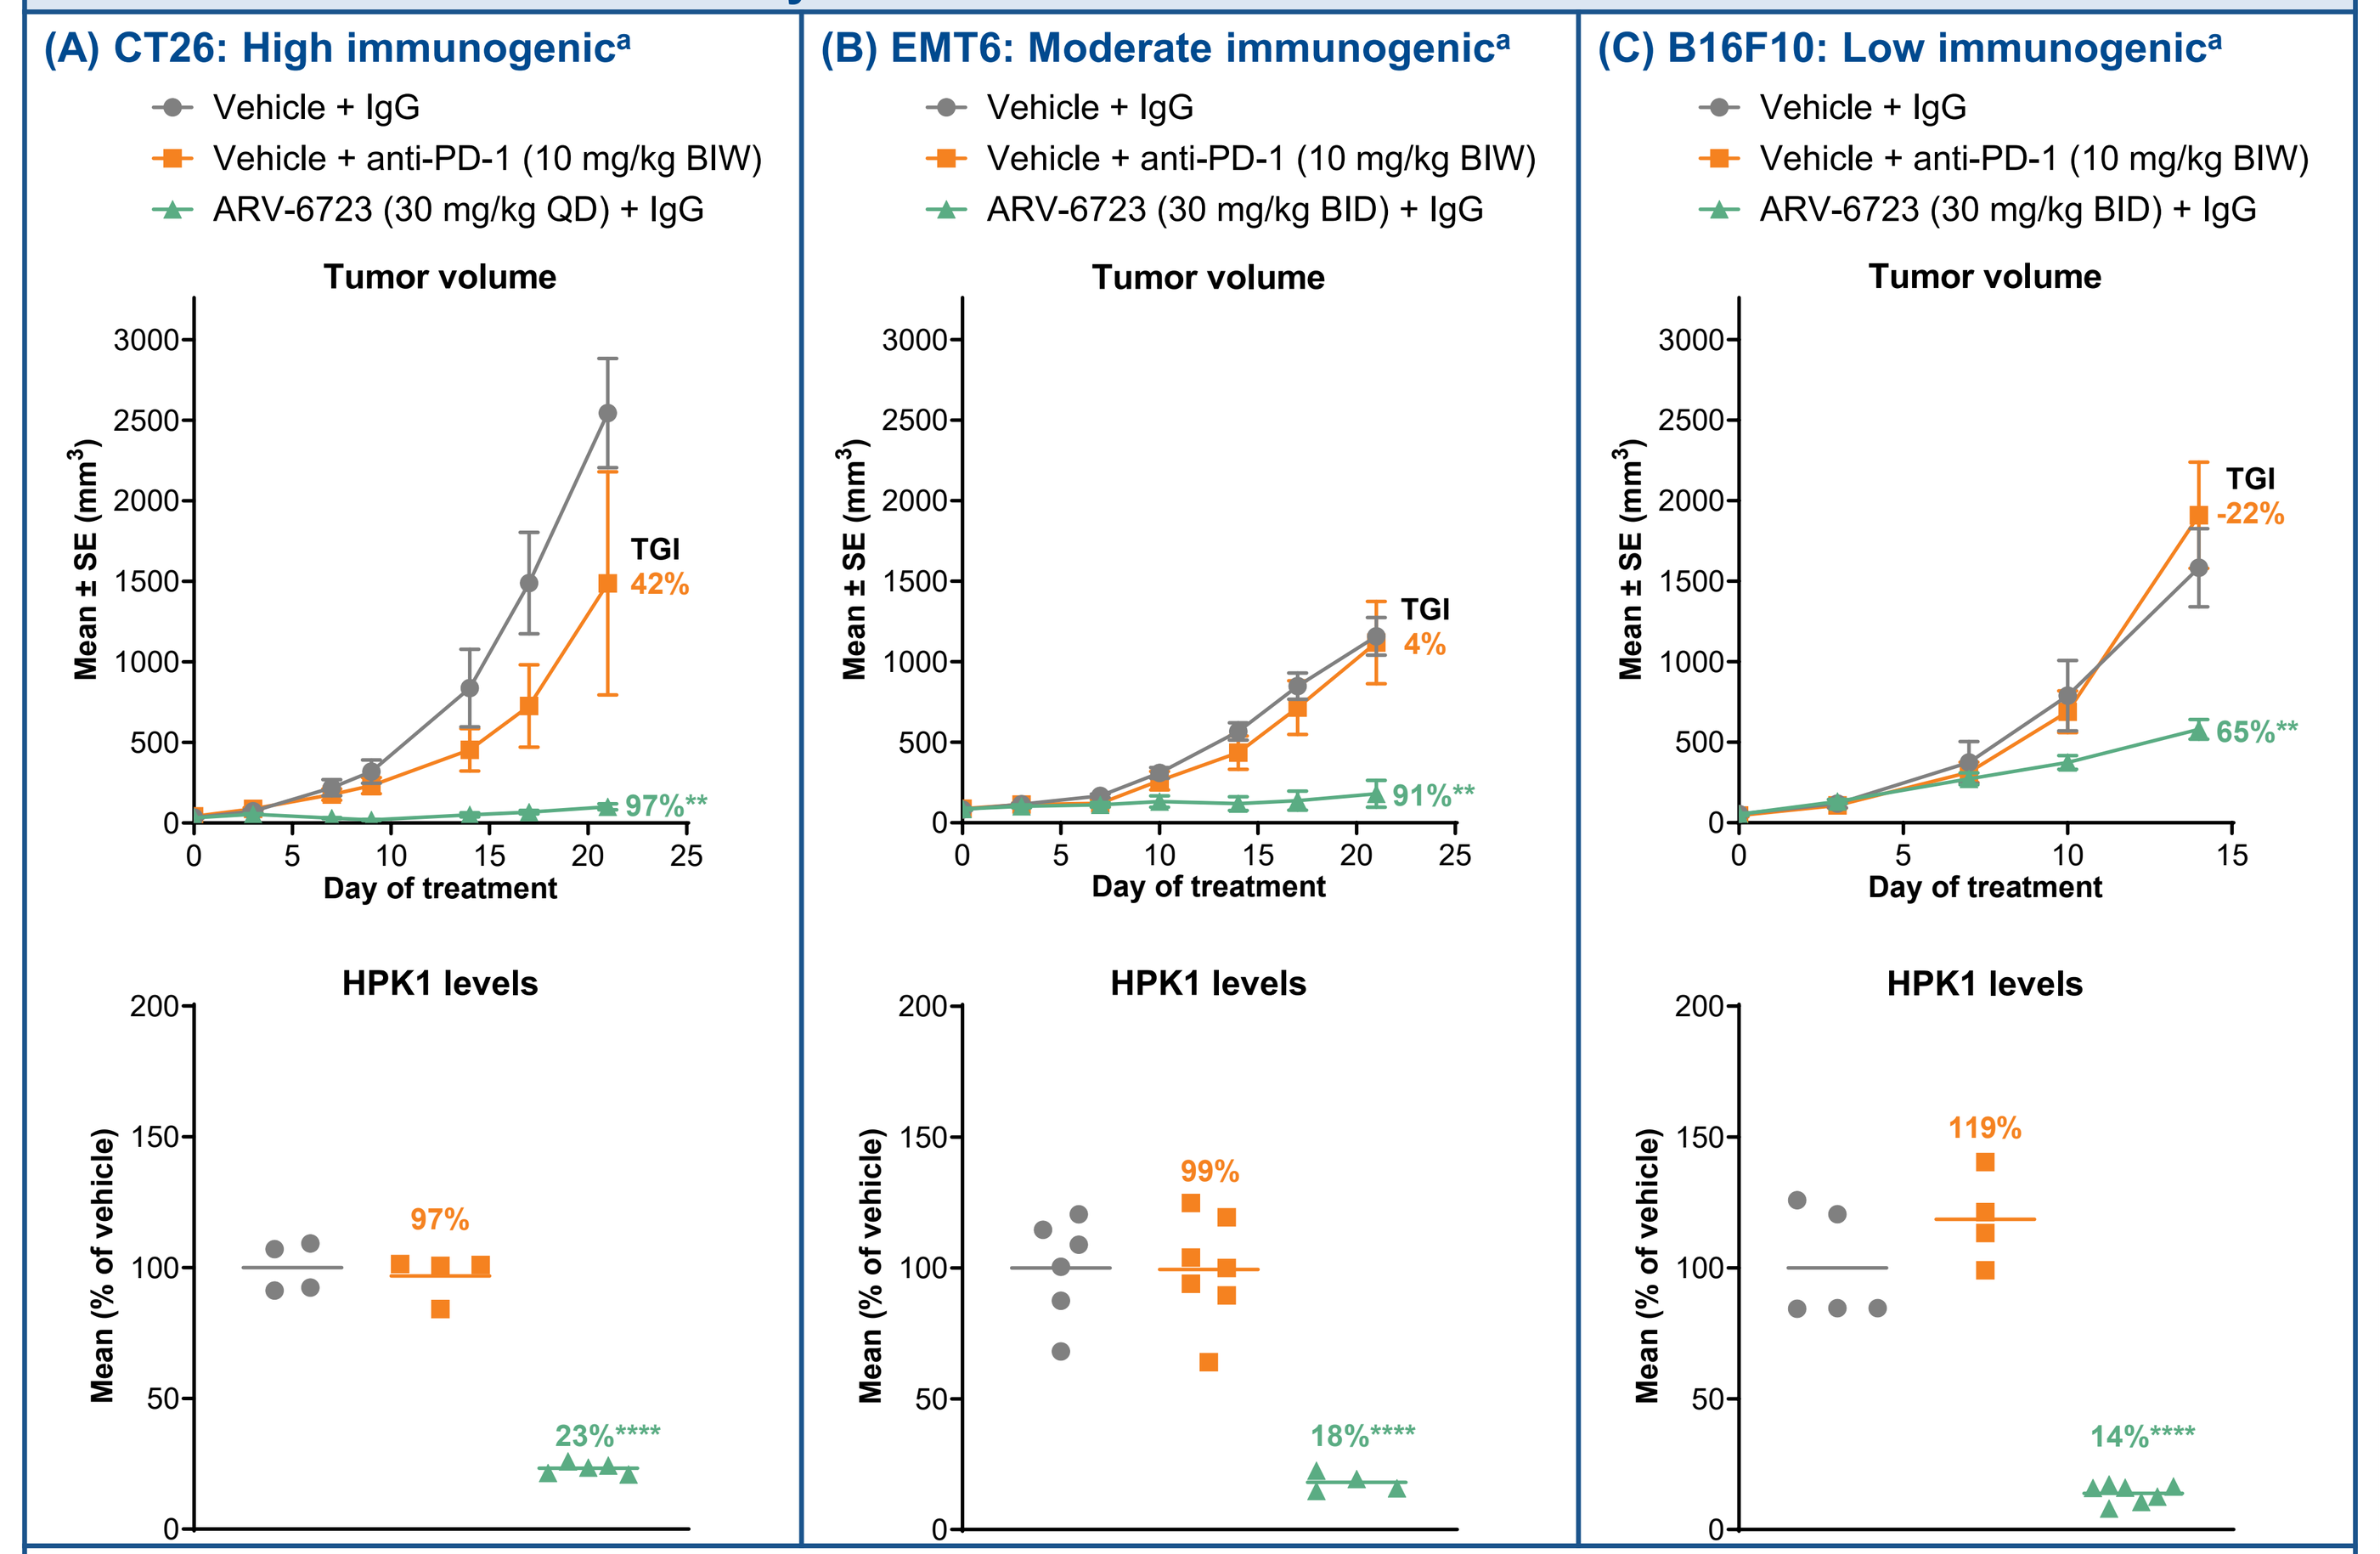

(A) CT26: High immunogenicᵃ
Vehicle + IgG
Vehicle + anti-PD-1 (10 mg/kg BIW)
ARV-6723 (30 mg/kg QD) + IgG
Tumor volume
TGI 42%
97%**
HPK1 levels
97%
23%****
(B) EMT6: Moderate immunogenicᵃ
Vehicle + IgG
Vehicle + anti-PD-1 (10 mg/kg BIW)
ARV-6723 (30 mg/kg BID) + IgG
Tumor volume
TGI 4%
91%**
HPK1 levels
99%
18%****
(C) B16F10: Low immunogenicᵃ
Vehicle + IgG
Vehicle + anti-PD-1 (10 mg/kg BIW)
ARV-6723 (30 mg/kg BID) + IgG
Tumor volume
TGI -22%
65%**
HPK1 levels
119%
14%****
Mean ± SE (mm³)
Day of treatment
Mean (% of vehicle)

Tumor volume and spleen HPK1 levels in (**A**) BALB/c mice implanted with CT26 tumor cells, (**B**) BALB/c mice with EMT6 tumors, and (**C**) C57BL/6 mice with B16F10 tumors. In each experiment, mice were implanted subcutaneously with the respective tumor line, tumors were established, and mice were randomized to treatment. Terminal (day 14-21) HPK1 protein levels were normalized to β-actin levels and quantified by Western blot. If tumor volume surpassed the ethical threshold, the last measured value was extended to end of study. *P*-values were calculated using one-way ANOVA with Dunnett's multiple comparisons (mean vs vehicle). $^{**}P<0.01$; $^{****}P<0.0001$.
ᵃTumors are classified as high vs low immunogenic based on differential responses to immune checkpoint blockade and other immunotherapies.[14]
ANOVA=analysis of variance; BID=twice daily; BIW=twice weekly; HPK1=hematopoietic progenitor kinase 1; IgG=immunoglobulin; PD-1=programmed cell death protein 1; QD=once daily; SE=standard error; TGI=tumor growth inhibition.

Figure 3: TGI, HPK1 degradation, pSLP76, and cell-population analysis in a low-immunogenic tumor model (B16F10) treated with ARV-6723 vs an investigational HPK1i

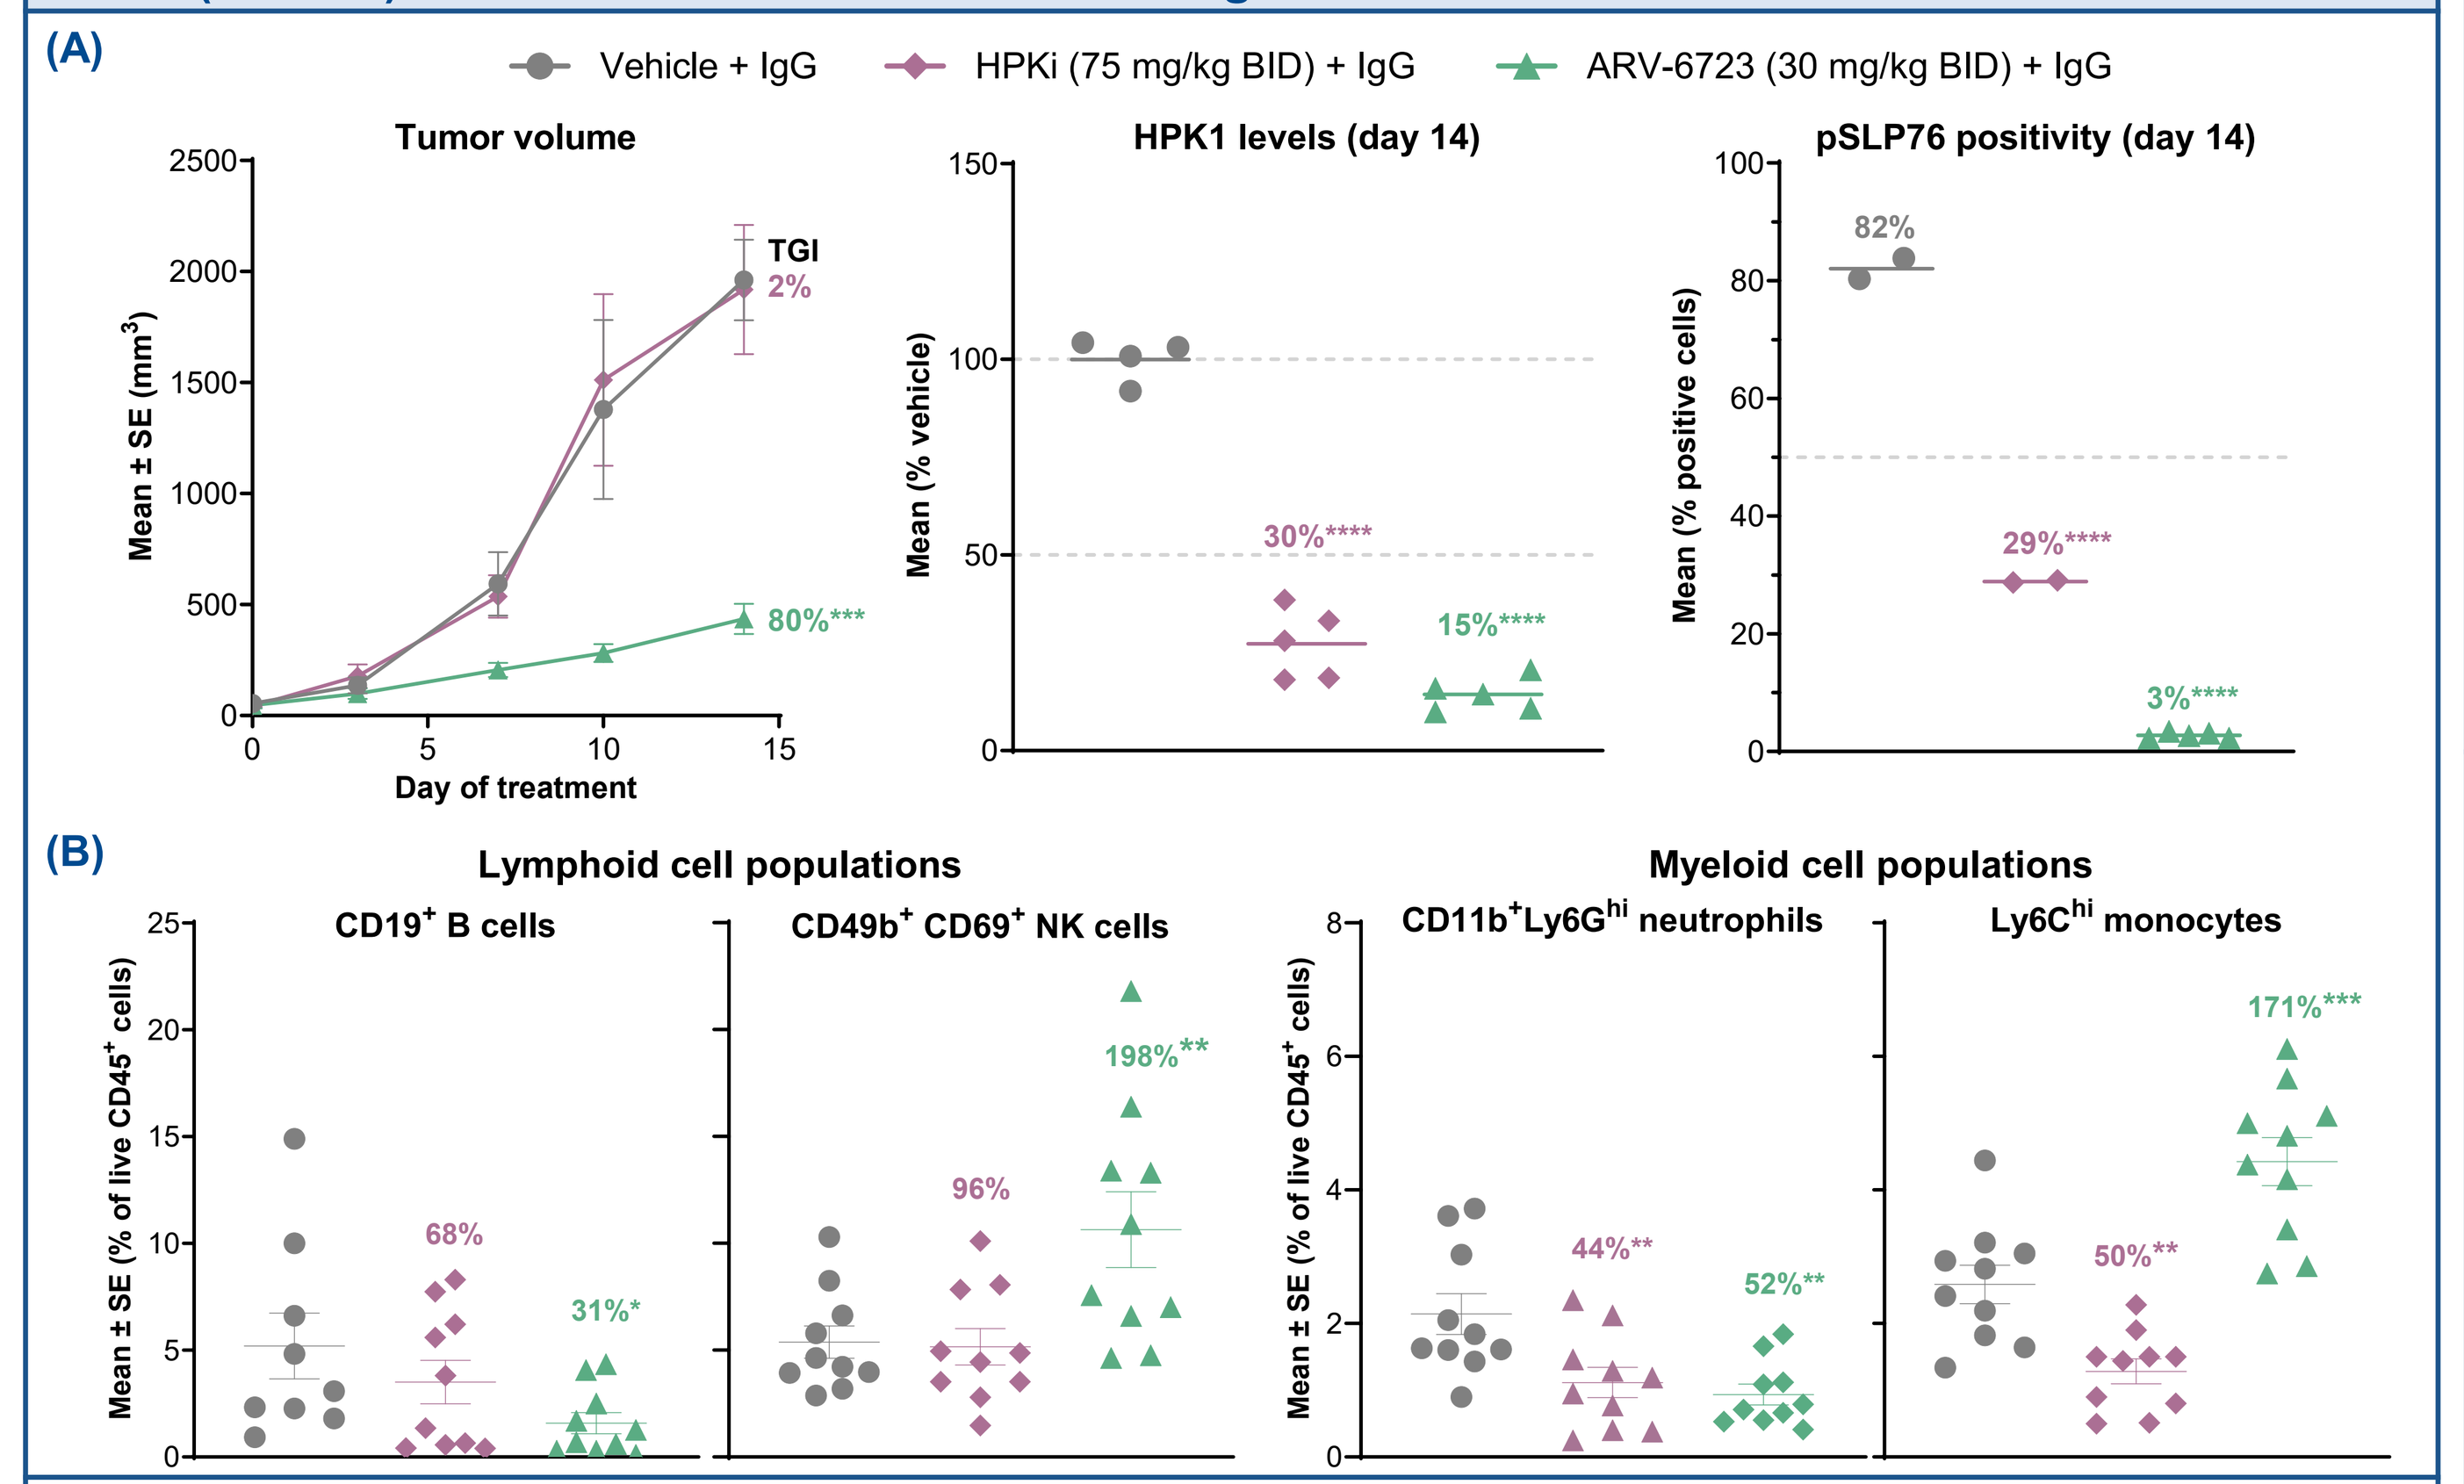

(A)
Vehicle + IgG
HPKi (75 mg/kg BID) + IgG
ARV-6723 (30 mg/kg BID) + IgG
Tumor volume
TGI 2%
80%***
HPK1 levels (day 14)
30%****
15%****
pSLP76 positivity (day 14)
82%
29%****
3%****
(B)
Lymphoid cell populations
CD19⁺ B cells
68%
31%*
CD49b⁺ CD69⁺ NK cells
96%
198%**
Myeloid cell populations
CD11b⁺Ly6Gʰⁱ neutrophils
44%**
52%**
Ly6Cʰⁱ monocytes
50%**
171%***

C57BL/6 mice were implanted subcutaneously with the B16F10 tumor line, tumors were given time to establish, and mice were randomized to the following treatments: vehicle (PO QD + IgG2a 10 mg/kg IP BIW); ARV-6723 (30 mg/kg PO QD + IgG2a 10 mg/kg IP BIW); or an investigational HPK1i (75 mg/kg PO QD + IgG2a 10 mg/kg IP BIW).
(**A**) Tumor volume, spleen HPK1 levels, and pSLP76 positivity were assessed after 14 days of treatment. (**B**) A separate cohort of mice received treatment until day 7, at which point tumors were harvested, dissociated, and immunophenotyped by flow cytometry. Lymphoid and myeloid cell populations were quantified. Percentage values shown are the percent difference compared with the vehicle group. *P*-values were determined by a one-way ANOVA with Dunnett's multiple comparisons (mean vs vehicle).
$^{*}P<0.05$; $^{**}P<0.01$; $^{***}P<0.005$; $^{****}P<0.0001$.
ANOVA=analysis of variance; BIW=twice weekly; HPK1=hematopoietic progenitor kinase 1; HPK1i=HPK1 inhibitor; IgG=immunoglobulin; IgG2a=immunoglobulin G2a; IP=intraperitoneal; NK=natural killer; PO=oral; QD=once daily; TGI=tumor growth inhibition; SE=standard error.

Figure 4: TGI, HPK1 degradation, pSLP76, and cell-population analysis in a high-immunogenic tumor model (CT26) treated with ARV-6723 vs an investigational HPK1i

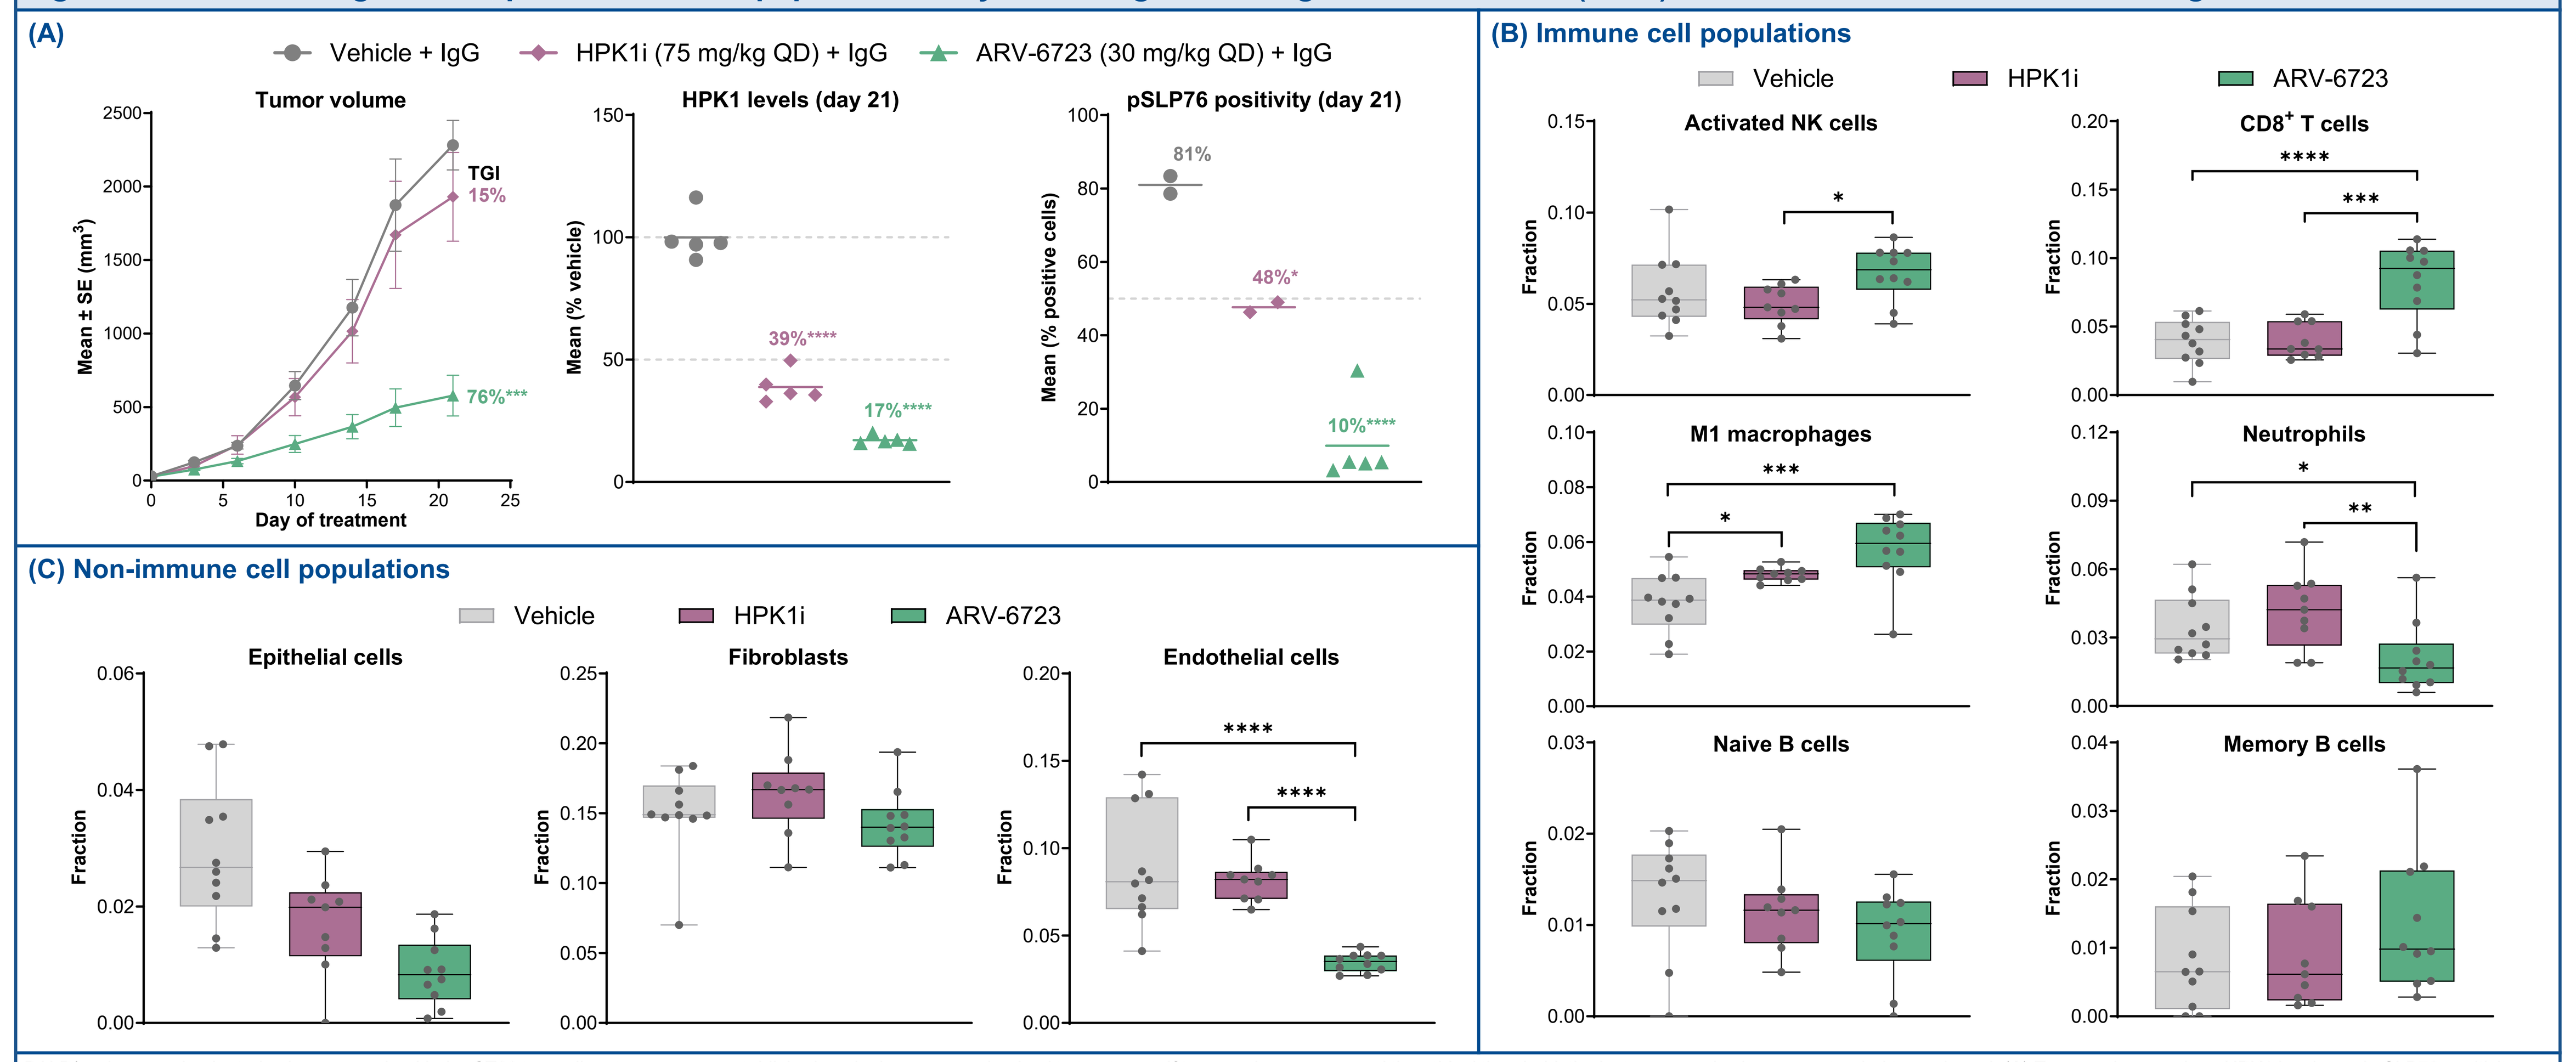

(A)
Vehicle + IgG
HPK1i (75 mg/kg QD) + IgG
ARV-6723 (30 mg/kg QD) + IgG
Tumor volume
TGI 15%
76%***
HPK1 levels (day 21)
39%****
17%****
pSLP76 positivity (day 21)
81%
48%*
10%****
(B) Immune cell populations
Vehicle
HPK1i
ARV-6723
Activated NK cells
CD8⁺ T cells
M1 macrophages
Neutrophils
Naive B cells
Memory B cells
(C) Non-immune cell populations
Epithelial cells
Fibroblasts
Endothelial cells
Fraction

BALB/c mice were implanted subcutaneously with the CT26 tumor line, tumors were established, and mice were randomized to treatment. If tumor volume surpassed the ethical threshold, the last measured value was extended to study end. (**A**) Tumor volume, spleen HPK1 levels, and pSLP76 positivity were measured. Terminal (day 21) HPK1 protein levels were normalized to β-actin levels and quantified by Western blot. pSLP76 percent positivity was evaluated with ex vivo stimulation using cells collected from the tumor draining lymph node on day 21; percentage of positive cells was quantified by flow cytometry. *P*-values in panel **A** are from a one-way ANOVA with Dunnett's multiple comparisons (each mean vs vehicle). (**B-C**) Mice received treatment until day 10, at which point tumors were harvested, and RNA was extracted and sequenced via bulk RNA-seq. Specific cell population proportions were inferred by iSort™ Fractions (CiberMed) for (**B**) immune and (**C**) non-immune cell types. Cell fractions were fitted with beta regression. Pairwise contrasts between group-estimated marginal means were tested using a Wald z-test, and *P*-values were adjusted for multiple comparisons using Tukey's method.
$^{*}P<0.05$; $^{**}P<0.01$; $^{***}P<0.005$; $^{****}P<0.0001$.
ANOVA=analysis of variance; HPK1=hematopoietic progenitor kinase 1; HPK1i=HPK1 inhibitor; IgG=immunoglobulin; NK=natural killer; pSLP76=phosphorylated SH2 domain-containing leukocyte protein of 76 kDa; RNA-seq=RNA sequencing; SE=standard error; TGI=tumor growth inhibition.

Figure 5: Differential expression of upregulated genes and pathway analysis in high- and low-immunogenic tumor models treated with ARV-6723 vs an investigational HPK1i

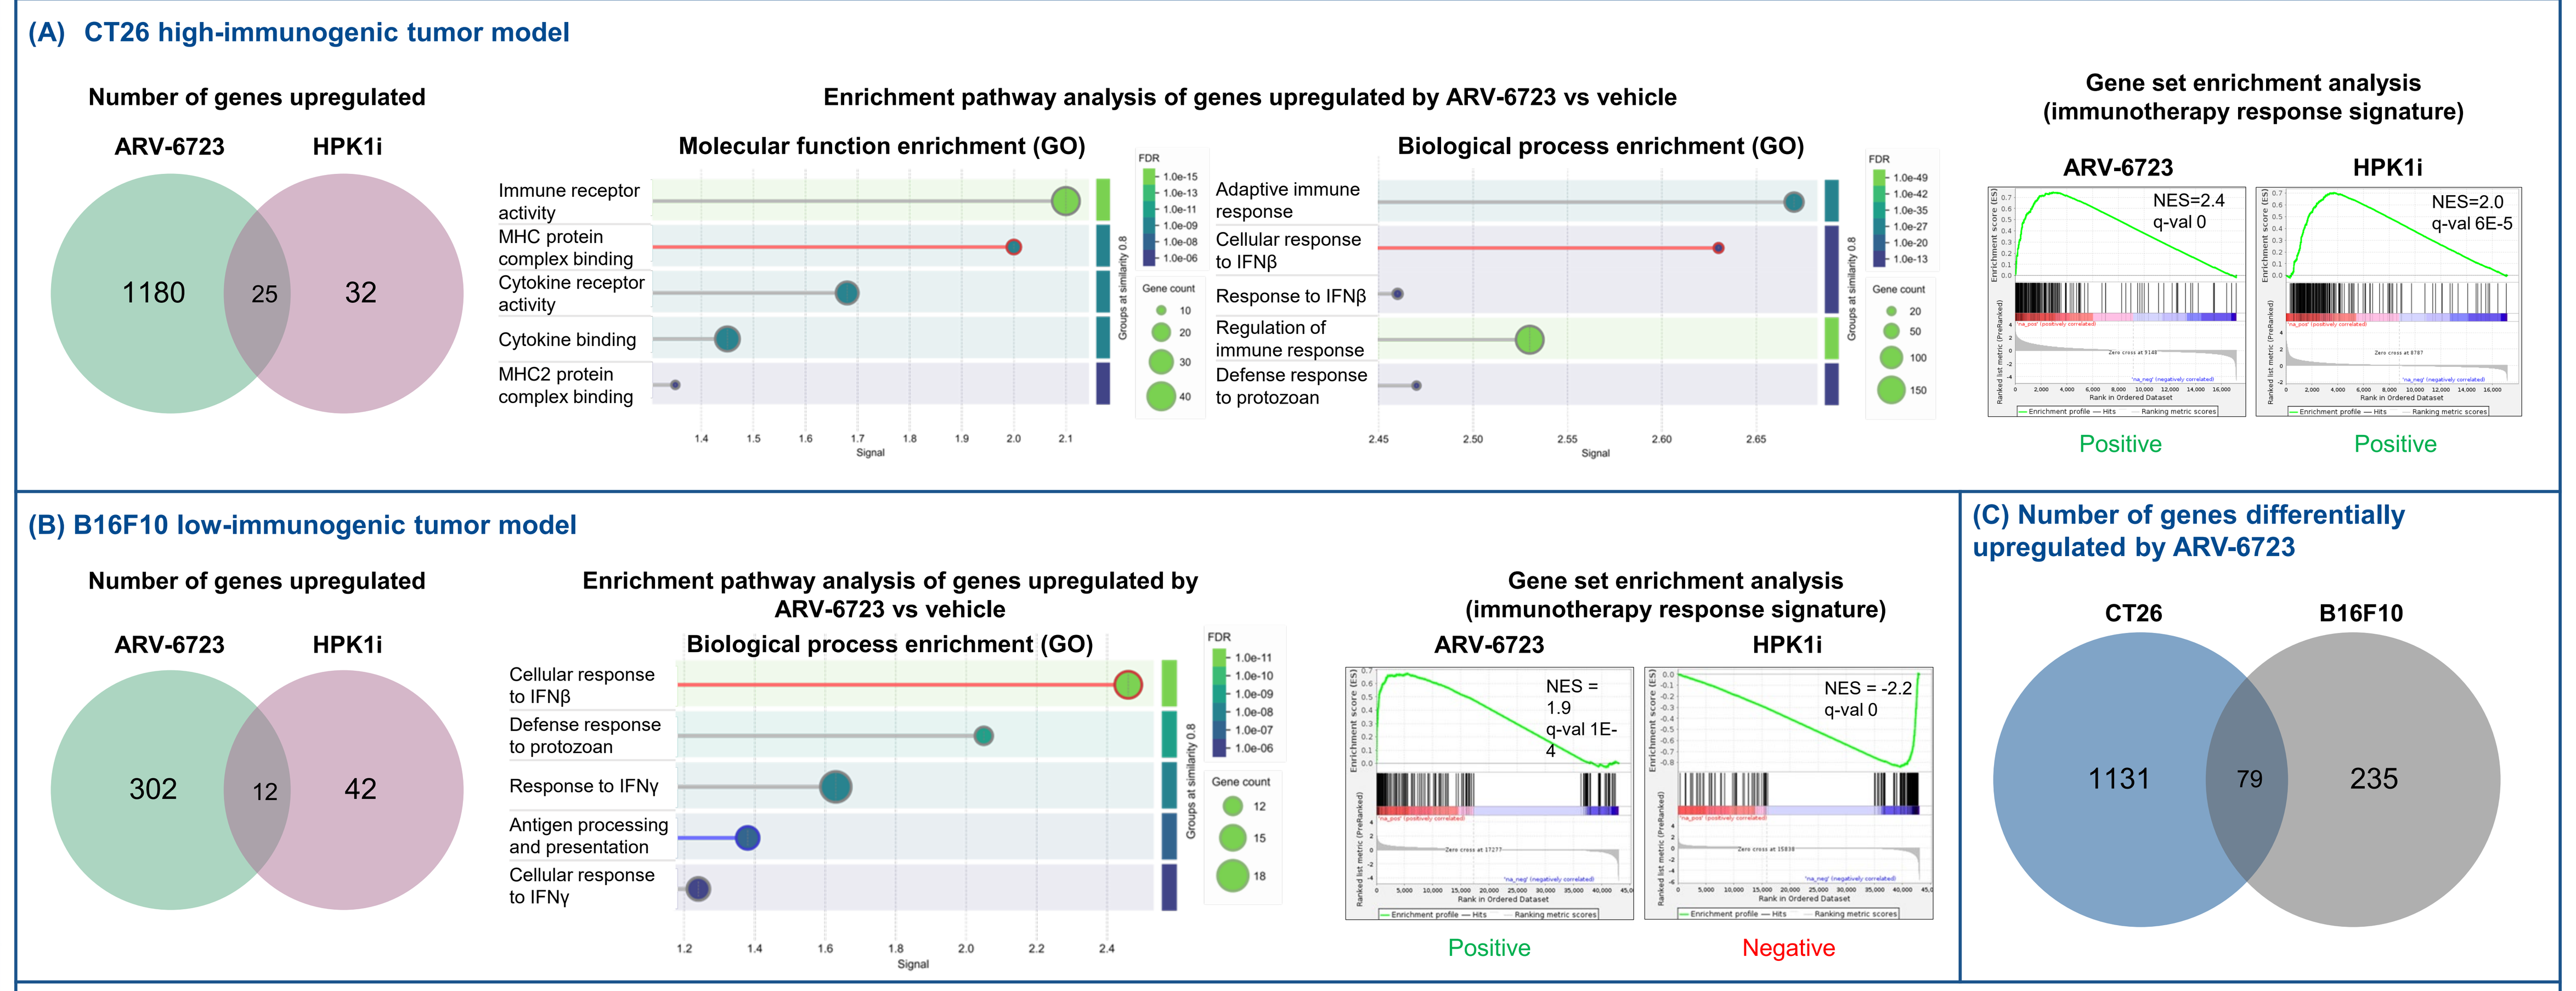

(A) CT26 high-immunogenic tumor model
Number of genes upregulated
ARV-6723
HPK1i
1180
25
32
Enrichment pathway analysis of genes upregulated by ARV-6723 vs vehicle
Molecular function enrichment (GO)
Immune receptor activity
MHC protein complex binding
Cytokine receptor activity
Cytokine binding
MHC2 protein complex binding
Biological process enrichment (GO)
Adaptive immune response
Cellular response to IFNβ
Response to IFNβ
Regulation of immune response
Defense response to protozoan
Gene set enrichment analysis (immunotherapy response signature)
ARV-6723
NES=2.4
q-val 0
HPK1i
NES=2.0
q-val 6E-5
Positive
Positive
(B) B16F10 low-immunogenic tumor model
Number of genes upregulated
ARV-6723
HPK1i
302
12
42
Enrichment pathway analysis of genes upregulated by ARV-6723 vs vehicle
Biological process enrichment (GO)
Cellular response to IFNβ
Defense response to protozoan
Response to IFNγ
Antigen processing and presentation
Cellular response to IFNγ
Gene set enrichment analysis (immunotherapy response signature)
ARV-6723
NES = 1.9
q-val 1E-4
HPK1i
NES = -2.2
q-val 0
Positive
Negative
(C) Number of genes differentially upregulated by ARV-6723
CT26
B16F10
1131
79
235

Bulk RNA-seq data were processed using Nextflow nf-core/rnaseq pipeline. Raw sequencing reads were aligned to the mouse reference genome by STAR and gene expression was quantified by RSEM using default parameters. Differential expression analyses were performed using R Bioconductor package DESeq2. Adjusted *P*-values <0.05 and log2FC values >1 were used to identify genes upregulated by ARV-6723 or HPK1i as compared to vehicle. Pathway enrichment was assessed by over-representation analysis for ARV-6723 upregulated genes using GO Molecular Function and Biological Process databases, and pre-ranked GSEA for ARV-6723 and HPK1i using gene sets obtained from the mouse MSigDB. Results for the high-immunogenic CT26 tumor model and low-immunogenic B16F10 tumor model are displayed in panels **A** and **B**, respectively. The number of ARV-6723-upregulated genes that were commonly and uniquely upregulated in the 2 tumor models are displayed in panel **C**.
FDR=false discovery rate; GO=Gene Ontology; GSEA=gene set enrichment analysis; HPK1i=hematopoietic progenitor kinase 1 inhibitor; IFNβ= interferon beta; IFNγ=interferon gamma; MHC2=major histocompatibility complex class 2; MSigDB=Molecular Signatures Database; NES=normalized enrichment score; RNA-seq=RNA sequencing.

Figure 6: Activity in a low-immunogenic pancreatic cancer model (KPCY-2699) treated with ARV-6723, an anti-PD-1 antibody, or an investigational HPK1i

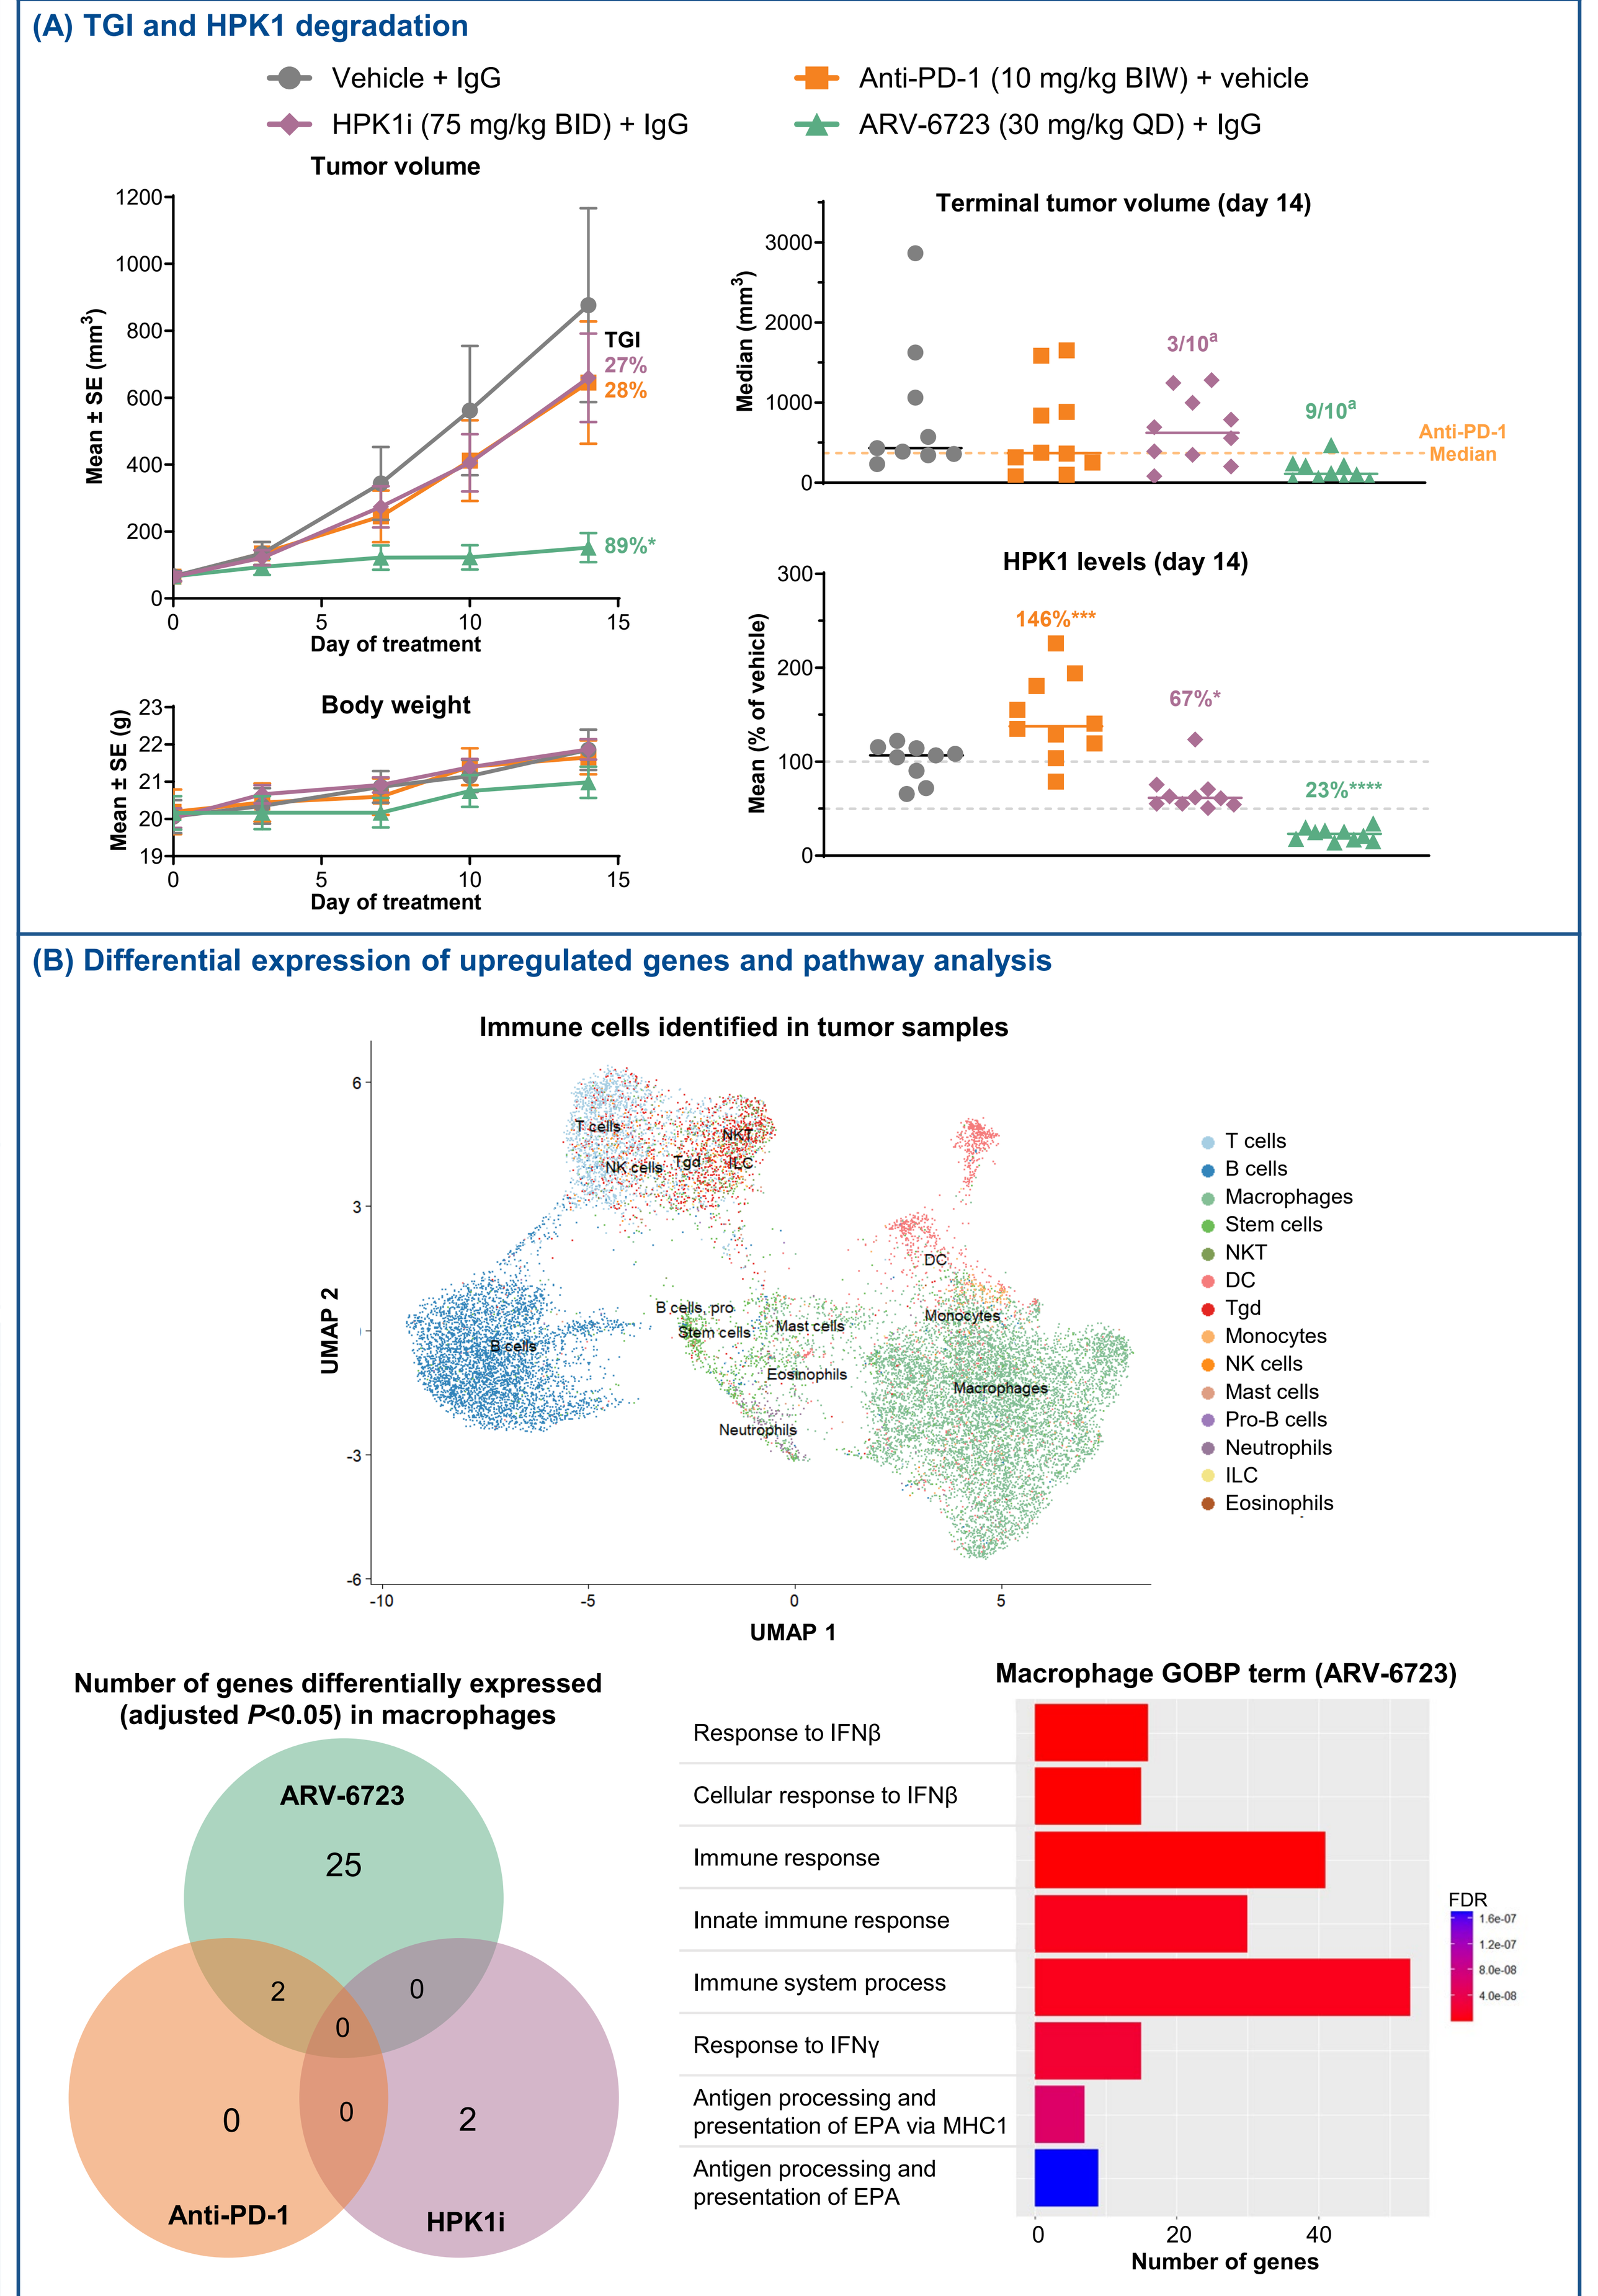

(A) TGI and HPK1 degradation
Vehicle + IgG
HPK1i (75 mg/kg BID) + IgG
Anti-PD-1 (10 mg/kg BIW) + vehicle
ARV-6723 (30 mg/kg QD) + IgG
Tumor volume
TGI 27% 28%
89%*
Body weight
Terminal tumor volume (day 14)
3/10ᵃ
9/10ᵃ
Anti-PD-1 Median
HPK1 levels (day 14)
146%***
67%*
23%****
(B) Differential expression of upregulated genes and pathway analysis
Immune cells identified in tumor samples
UMAP 1
UMAP 2
Number of genes differentially expressed (adjusted P<0.05) in macrophages
ARV-6723
Anti-PD-1
HPK1i
25
2
0
0
0
0
2
Macrophage GOBP term (ARV-6723)
Response to IFNβ
Cellular response to IFNβ
Immune response
Innate immune response
Immune system process
Response to IFNγ
Antigen processing and presentation of EPA via MHC1
Antigen processing and presentation of EPA
Number of genes

(**A**) Tumor volume, body weight and HPK1 levels in spleen lysates from C57BL/6 mice implanted subcutaneously with KPCY clone #2699 and dosed with the specified treatment during a 14-day study (n=8–10 per group). HPK1 protein levels were normalized to β-actin levels and quantified by Western blot.
P-values were calculated using a one-way ANOVA with Dunnett's multiple comparisons (mean vs vehicle). $^{*}P<0.05$; $^{**}P<0.01$; $^{***}P<0.005$; $^{****}P<0.0001$.
(**B**) Single-nuclear RNA-seq data were processed using Cell Ranger with default parameters. The R package Seurat was used for further analysis. Low-quality nuclei with <500 quantified genes, >20% mitochondrial reads, together with potential doublets/multiplets were filtered out. Cell types were annotated using the ImmGen reference dataset. Differentially expressed genes in treatment vs vehicle in each immune cell type were identified using pseudobulk values and DESeq2. Pathway enrichment with ARV-6723 in macrophages was assessed using increased genes (FC>1.5, *P*<0.05) by overrepresentation analysis using the GO Biological Process database. Enriched processes are ranked by significance.
ᵃNumbers indicate the proportion of tumors below the median tumor volume in the anti-PD-1 group.
ANOVA=analysis of variance; BID=twice daily; BIW=twice weekly; DC=dendritic cell; EPA=endogenous peptide antigen; FC=fold change; FDR=false discovery rate; GO=Gene Ontology; GOBP=Gene Ontology Biological Process; HPK1=hematopoietic progenitor kinase 1; HPK1i=HPK1 inhibitor; IFNβ=interferon beta; IFNγ=interferon gamma; IgG=immunoglobulin; ILC=innate lymphoid cell; MHC1=major histocompatibility complex class I; NK=natural killer; NKT=natural killer T cell; PD-1=programmed cell death protein 1; QD=once daily; RNA-seq=RNA sequencing; SE=standard error; Tgd=gamma delta T cell; TGI=tumor growth inhibition; UMAP=uniform manifold approximation and projection; UMI=unique molecular identifiers.